In this subject N°21, we can observe a reduction of superficial wrinkles and visible pores on the MELA BRIGHT® [C+] side, this reduction is not visible on the Hydroquinone 4% side.

T0 **T2 months** **T0** **T2 months**

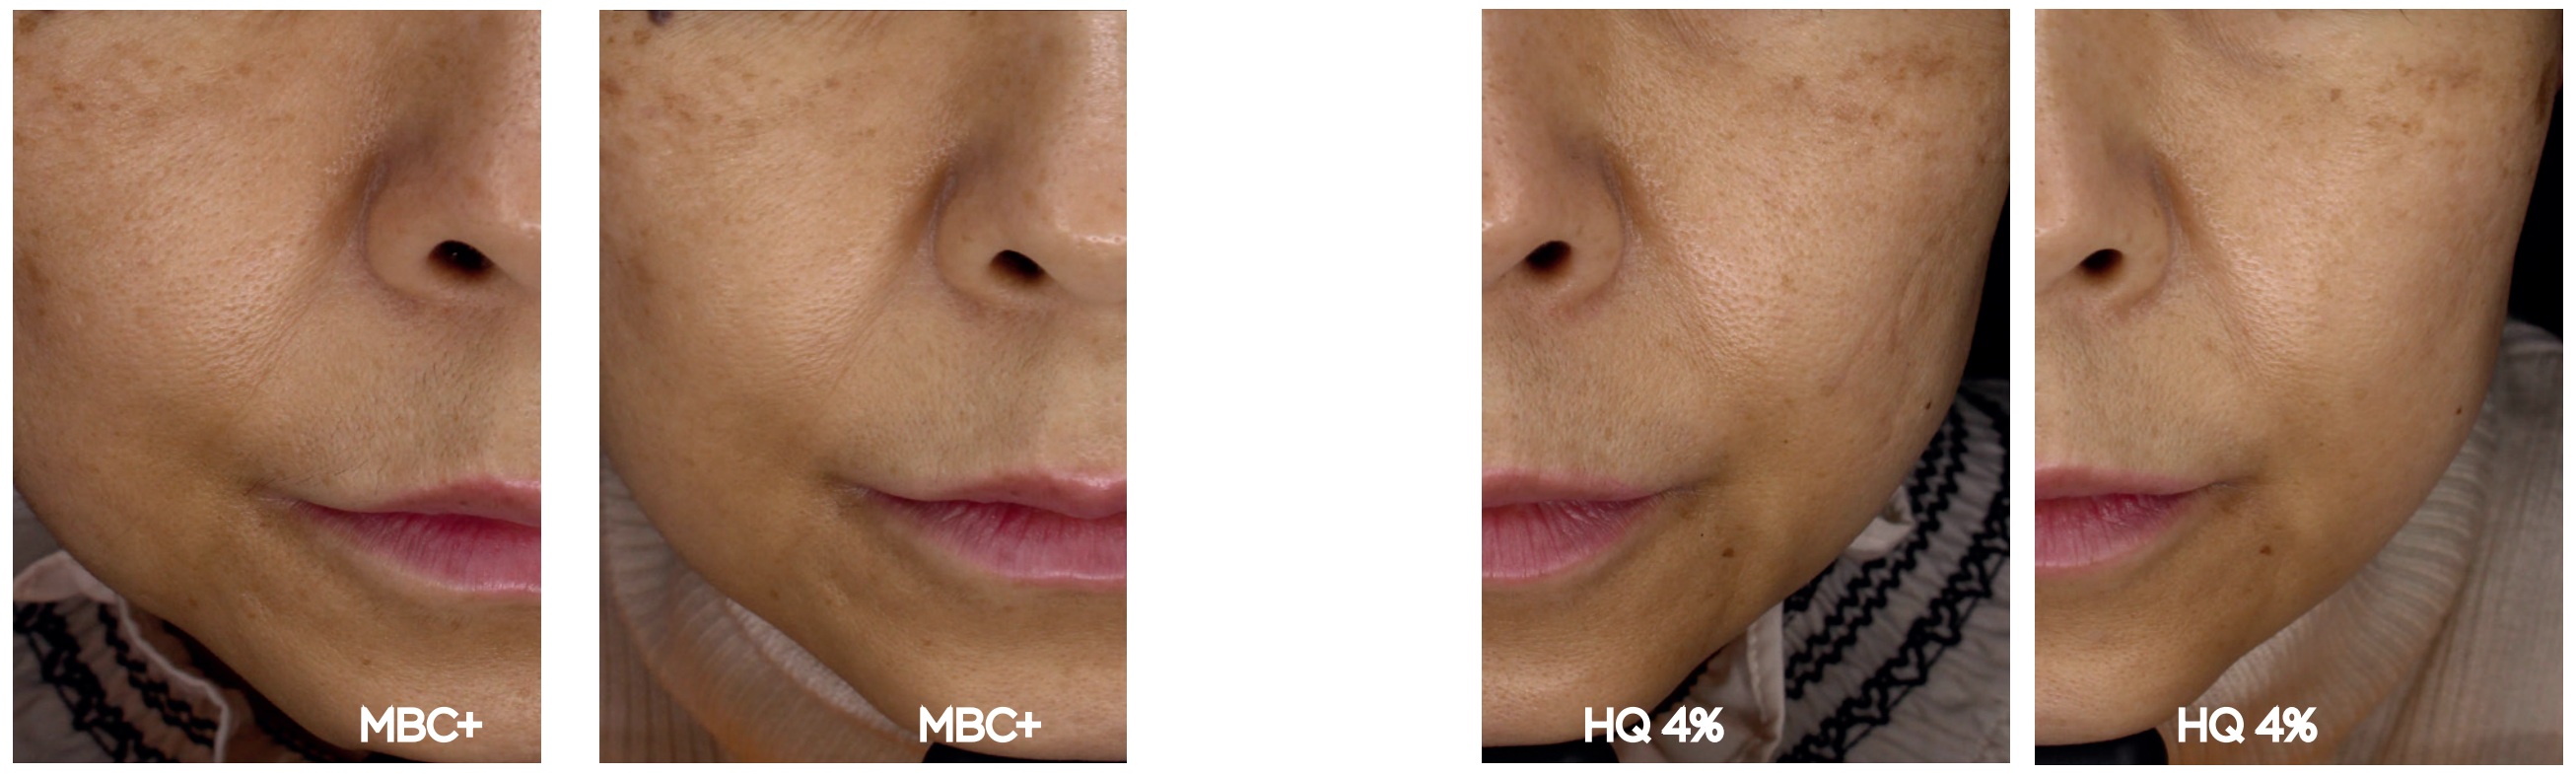
MBC+
MBC+
HQ 4%
HQ 4%

CONCLUSION

After 2 months of treatment, an average pigmentation decrease of 14% (visible light) and 17% (Wood lamp) was measured on the MELA BRIGHT® [C+] side, compared to an average pigmentation decrease of 15% (visible light) and 19% (Wood lamp) on the HYDROQUINONE 4% side.

We can therefore conclude that both treatments have similar efficacy on melasma after 2 months with an average severity decrease of 18% (Wood lamp)..
MELA BRIGHT® [C+] can consequently be a promising option to safely substitute hydroquinone treatments for phototypes I to VI or to safely use after hydroquinone treatment to manage residual pigmentation and also as a maintenance therapy for melasma, in first intention or in combination with cosmetic procedures. The patients expressed satisfaction with this hyperpigmentation treatment and their skin looked glowier, texture improved, pores visibly reduced and younger.

Conflicts of interest Alfred Marchal PhD is employee of Alphascience Research, Carmen Domínguez, PharmD. is employee of Alphascience.

REFERENCES

1 Handel et al. Melasma: a clinical and epidemiological review. Anais brasileiros de dermatologia, 89(5), 771–782, 2014.

2 Juliano et al. «Spreading of Dangerous Skin-Lightening Products as a Result of Colourism: A Review» Applied Sciences 12, no. 6: 3177, 2022.

3 Baumann et al., "Skincare and nonsurgical skin rejuvenation" Plastic surgery, Vol 2: Aesthetic surgery, 4, 23-37

4 Cestari et al. Acquired hyperpigmentations. Anais brasileiros de dermatologia, 89(1), 11–25, 2014.

5 Arellano et al. Guias de diagnóstico y manejo de melasma. Dermatología CMQ. 2017;16(1):12-23.

6 Kwon, et al. Melasma: Updates and perspectives. Exp Dermatol. 2019; 28: 704– 708.

7 Sanchez et al. Melasma: a clinical, light microscopic, ultrastructural, and immunofluorescence study. J Am Acad Dermatol. 1981;4(6):698-710. doi:10.1016/s0190-9622(81)70071-9

8 Filoni et al. Melasma: How hormones can modulate skin pigmentation. J Cosmet Dematol. 2019;1-6.

9 Zhou et al. Phytic Acid in Health and Disease. Critical Reviews in Food Science and Nutrition. 1995; 35: 495-508

10 Rendon et al - Review of Skin-Lightening Agents - Dermatologic Surgery.

11 Glycine derivative capable of inhibiting melanin formation and composition using the same (US 8,927,499 B2) https://www.google.com/patents/US8927499

12 Ražná et al. Properties of Ginkgo biloba L.: Antioxidant Characterization, Antimicrobial Activities, and Genomic MicroRNA Based Marker Fingerprints. International journal of molecular sciences, 21(9), 3087, 2020.

13 Gallego-Villar et al., Cysteamine revisited: repair of arginine to cysteine mutations. J Inherit Metab Dis, 40 (4), 555-567, 2017.

14 Desai et al. Topical Stabilized Cysteamine as a New Treatment for Hyperpigmentation Disorders: Melasma, Post-Inflammatory Hyperpigmentation, and Lentigines. J Drugs Dermatol. 2021, 20(12), 1276-1279, 2021.

14 Chavin et al., Some potent melanin depigmentary agents in the black goldfish. Naturwissenschaften.53(16), 413-414, 1966.

15 Ito et al., Depigmentation of black guinea pig skin by topical application of cysteaminylphenol, cysteinylphenol and related compounds. J Invest Dermatol, 88(1), 77-82, 1987.

16 Parvez et al., Survey and mechanism of skin depigmenting and lightening agents. Phytother Res, 20(11), 921-934, 2006

17 Qiu et al. Inhibition of melanin synthesis by cystamine in human melanoma cells. J Invest Dermatol 2000; 114:21–7

18 De Matos et al., Effect of cysteamine on gluthatione level and development capacity of bovine oocyte matured in vitro. Mol Reprod Dev, 42(4), 432-436, 1995.

19 Sakurai et al., Studies on the sulfur-containing chelating agents. XXXI. Catalytic effect of copper (II) ion to formation mixed disulfite. Chem Pharm Bull, 19(7), 1416-1423, 1971.

20 Bottu et al., The effect of quenchers on the chemiluminescence of luminol and lucigenin. J Biolumin Chemilumi, 3(2), 59-65, 1989

21 Gillbro et al., The melanogenesis and mechanisms of skincare lightening agents-existing and new approaches. Int J Cosm Sci, 33 (3), 210-221, 2011

22 Stratigos et al., Optimal management of recalcitrant disorders of hyperpigmentation in dark-skinned patients.. Am J Clin Dermatol, 5 (3), 161-168, 2004.

23 Gordon et al. Interleukin-6 receptor antagonists in critically ill patients with Covid-19.N Engl J Med 384: 1491–1502,2021.

24 Okamura et al., Cysteamine modulates oxidative stress and blocks myofibroblast activity in CKD.J Am Soc Nephrol, 25: 43–54, 2014.

25 Jeitner et al., Cystamine and cysteamine as inhibitors of transglutaminase activity in vivo. Biosci. Rep,38: BSR20180691, 2018.

«MELASMA: A HEMIFACIAL STUDY ON A CYSTEAMINE SERUM AS AN ALTERNATIVE TO HYDROQUINONE »

Aoi Nakano, Dermatologist, AOI Clinic, Tokyo
Alfred Marchal PhD, Organic Chemistry
Carmen Domínguez, PharmD

AMWC 2023
21ST AESTHETIC & ANTI-AGING MEDICINE WORLD CONGRESS
OFFICIAL E-POSTER

ABSTRACT

This hemifacial study is an evaluation of the depigmenting efficacy of an antioxidant serum with 3% cysteamine and 8% L-Ascorbic acid (MELA BRIGHT ®[C+] SERUM) versus a reference treatment (Hydroquinone 4%) in 24 patients with melasma during 4 months and preliminary results after 2 months. The preliminary 2 months clinical trial results reveal both treatments used have similar efficacy with an average severity decrease of 18% observed in melasma.

BACKGROUND

Hyperpigmentary disorders remain an important concern at dermatology and Aesthetic medicine visits worldwide. The prevalence varies depending on the location and ethnicity. These disorders are observed in people of all ethnic backgrounds and locations but the prevalence is higher for populations with darker Fitzpatrick phototypes, and more exposed to sunlight[1]. Several products with active ingredients that target different stages of melanogenesis, have arrived to the market to respond to this important need. Although different combination therapies have been proven effective, the majority of the cosmetic products available lack effectiveness, and the most effective first-line proven treatment, have prompted several concerns about the safety, lead to strict regulations and bans in several countries[2] and more recently as an OTC in the US in September 2020. The development of exogenous ochronosis, worsened hyperpigmentation, occular reactions and other side effects including nail discoloration and contact dermatitis, along with the counter-indication on darker phototypes, explains the existing need for a safer and effective product[3].

MELASMA

Melasma is an acquired and chronic pigmentary disorder that predominantly affects women. a symmetric acquired hypermelanosis, with stains in shades of brown to bluish gray, with irregular borders and located in more photo-exposed areas. It usually affects the face and neck[4]. This pigmentary disorder appears in all skin phototypes, especially in phototypes III to IV, more frequently in people living in areas subjected to intense ultraviolet radiation[1,5].Melasma is a disease of melanocytes that has an aging component, with features of photoaging[6]. The pathogenesis of melasma are not fully understood nowadays, but studies have shown that factors like genetic predisposition, hormones, photosensitizing medications and exposome (chronic UV exposure) play a role in the development. [6]. The management of melasma is highly challenging, because it is prone to frequent relapses despite successful clearance[7]. Pregnancies and oral contraceptives seem to stimulate melanogenesis, by inducing the synthesis of melanogenic enzymes such as tyrosinase and tyrosinase-related proteins 1 and 2, oestrogen stimulates melanogenesis in human melanocytes in cultures [8] [7] Estrogen, progesterone, and melanocyte-stimulating hormone levels are normally increased during the third trimester of pregnancy and seem to be involved. Some epidemiological studies showed that other factors seem to be involved: inflammation, basal membrane alteration and the exposure to visible light and UV. Cytokines and Growth factors released from sebocytes seem to exert paracrine effects on epidermal melanocytes[6]. Patient education should include sun avoidance.
The most used ways to quantify the severity of melasma are the Melasma Area and Severity Index (MASI) index developped by Kimbrough-Green and the Wood´s light as a useful diagnostic tool to evaluate the extension of the pigmented areas [7].

TREATMENTS

Several agents have been proposed to impact in different stages of melanogenesis and some of their mode of action include: inhibition of melanin production and melanosome transfer, increased turnover of keratinocytes, anti-inflammatory and antioxidant effects. Different compounds or combination therapies for melasma have been proven effective.
First-line therapy includes effective topical therapies, mainly with Hydroquinone (HQ) used solo or combined with other actives such as Tretinoine and Fluocinolone acetonide. HQ is the most widely used depigmenting agent but has an objectionable risk/benefit ratio safety profile[2]. Other topical treatments include kojic acid, azelaic acid, niacinamide, and resveratrol, among others exhibit depigmenting properties without severe adverse effects[6] but they don't allow to achieve the expected depigmenting results. Due to the therapeutic challenge associated with the pathophysiology of this hypermelanosis, the poor response to the majority of the treatments mentioned and the poorer safety profile of the first-line treatment, new depigmenting active ingredients and combinations have been proposed lately, like a pro-

mising new antioxidant serum with a combination of: 3% stable cysteamine, 8% stable Ascorbic acid, 1% Phytic acid, 1% acetyl Glycyl beta alanine, and 30% Ginkgo Biloba solution (MELA BRIGHT® [C+]).
Phytic acid is extracted from rice grain and has antioxidant, moisturizing, depigmenting, anticarcinogenic and sebum regulating properties [9].
L-ascorbic acid is one of the most potent topical antioxidants known. It has been proven to protect against collagen degradation. It blocks melanin transfer through a weakening action on dendrites, thus, has depigmenting properties[10]. It is also one of the most powerful existing antioxidants able to protect from oxidative factors of the exposome, responsible for the premature aging of the skin (UV, stress, pollution.) and DNA damage.
Acetyl glycyl β alanine is a peptide that exhibits a fast skin penetration, reducing melanin production and inhibiting its transference to keratinocytes. It inhibits stem cell factor and endothelin 1 which reduces the enzymes generating melanin (tyrosinase, proteins related to tyrosinases 1 and 2). It has been found to decrease the transcription factor associated with microphtalmia (MITF), linked to melanogenesis [11]
Gingko biloba is used in traditional medicine for its multiple benefits antioxidant, antibacterial, anti-fungal and vasoprotective properties. [12]

CYSTEAMINE

Cysteamine is an aminothiol naturally present in the body with an intrinsic antioxidant and it is known for its protective role. This molecule naturally occurs in the body due to the Coenzyme A degradation and is well distributed in the mammalian tissue and in human milk. [13, 14]
This thiol derivative discovered in 1953, has been extensively studied in the scientific literature and even though it has a good safety profile, due to the easy oxidation of this fragile molecule and the unpleasant smell when oxidised, this molecule wasn't used in cosmetics in recent years.
This powerful non-melanocytotoxic antioxidant molecule efficacy has been studied and compared to hydroquinone and modified Kligman's formulation. [14]
Cysteamine seems to be one of the most potent depigmenting agents available. This has been confirmed through several *in vivo* studies over the past few decades.[14, 15, 16] A study showed an *in vitro* 80% melanogenesis reduction. [17]
Even though, the depigmenting action of Cysteamine has been proven, the mechanisms are not fully understood. Thiol molecules are known to inhibit tyrosinase and peroxidase, essential enzymes in the melanogenesis pathway, leading to the conversion of tyrosine into a dopaquinone, and the polymerization of indoles into melanin. Thiols, seem to also increase levels of intracellular glutathione (GSH), slowing down the melanogenesis pathway and helping to restore the intracellular redox balance and amplify the natural depigmenting action. [18]
Cysteamine is a Copper and Iron chelating agent and a Dopaquinone scavenging agent. It seems that this molecule could slow down the conversion of tyrosine into dopaquinone by preventing Fenton-type reactions [18, 19]
This naturally present thiol derivative is a powerful antioxidant that helps fighting hydroxy free radicals and photooxidation, as such, it plays an important role in the melanogenesis pathway. [20, 21]
This depigmenting action seems also linked to its keratolytic effect, allowing an acceleration of the epidermal cell turnover that is prolonged due to the aging process and helping to get rid of the superficial epidermal layers that contain melanin and visibly look darker. [21, 22]
Its anti-inflammatory effect, mediated by the interruption of ROS-mediated inflammatory cascades and the inhibition of the proinflammatory protein transglutaminase 2, along with the other actions, make this an interesting and effective depigmenting active ingredient for inflammatory hyperpigmentation and other inflammatory pigmentary disorders. [23, 24, 25]

Stability

Due to its fragility, rapid oxidation and instability when not formulated correctly, it has suffered from different drawbacks during years, because of the strong odour and organoleptic properties when unstable.
It is very difficult to stabilize this molecule and this is why the general oxidation leads to a final 50% real concentration.
NextGen® Technology, creates a protecting shield by an ionization and electron transfer modulator effect, that allows a 93,2% concentration after 5 months in the bottle. This technology also improves the cysteamine stability inside the skin.

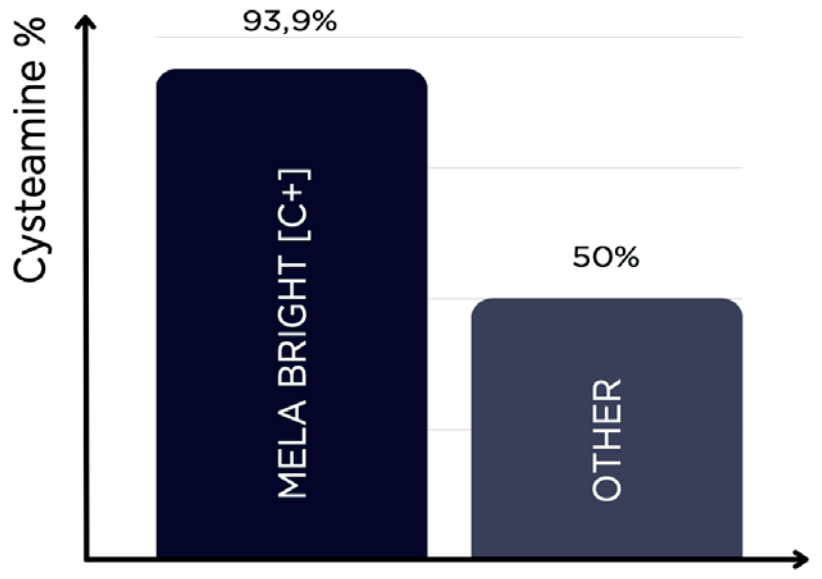
Cysteamine %
93,9%
MELA BRIGHT [C+]
50%
OTHER

Stability of Cysteamine measured with HPLC Spectroscopy by ALPHASCIENCE Research.

OBJECTIVE OF THE STUDY

The primary objective is to evaluate the effectiveness to improve severity in melasma of two topical depigmenting products: MELA BRIGHT® [C+] (ALPHASCIENCE) (3% Cysteamine + Acetyl-Glycyl-B-Alanine + 8% ascorbic acid + 2% phytic acid + ginkgo biloba) versus a product containing 4% Hydroquinone.
The secondary objective was to evaluate the skin tolerance and the general patient satisfaction.

METHODOLOGY

A prospective, randomized, open label trial was designed and the Research and Ethics Committee approved the protocol. They were recruited 24 melasma patients. As it was designed as a split face study, the patients were their controls. The study was designed in a three-phase modality for 4 months duration. For the first eight week patients were randomly assigned to application of the topical agent on half of the face, so MELA BRIGHT® [C+] (MBC+) was applied by gentle massage on the evenings and in the other half of the face the patient applied HYDROQUINONE 4% serum (HQ). In addition, all of the patients applied the same SPF 50 broad-spectrum sunscreen at morning.

Population and sample size:

24 melasma patients were included in our study, the age range was 25 to 60 years. All of the patients were evaluated through Wood's light and quantitative evaluation by Skin vision Visia Booth, so colorimetry, number and depth of wrinkles, pore size and texture of the skin as well as the calculated clinical age values were compared. MASI was calculated for melasma patients.
All of the patients answered a questionnaire for evaluation of the products applied and satisfaction. Basal (T0) and preliminary initial 8 weeks (T2) results were analysed.

Statistical Analysis:

The general data were analysed using descriptive statistics, mean and standard deviation for the quantitative variables with normal distribution, percentiles in the case of those that are not and frequencies and percentages for the categorical variables. It was considered that there was a statistically significant difference when the value of p was < 0.05.

RESULTS

PIGMENTATION INTENSITY

In the graph below we can observe the decrease of the pigmentation score at T0 and T2 months. An average pigmentation decrease of 14% (visible light) and 17% (Wood lamp) was measured on the MELA BRIGHT® [C+] side ($p<0.005$), compared to a decrease of 15% (visible light) and 19% (Wood lamp) on the HYDROQUINONE 4% side ($p<0.05$).

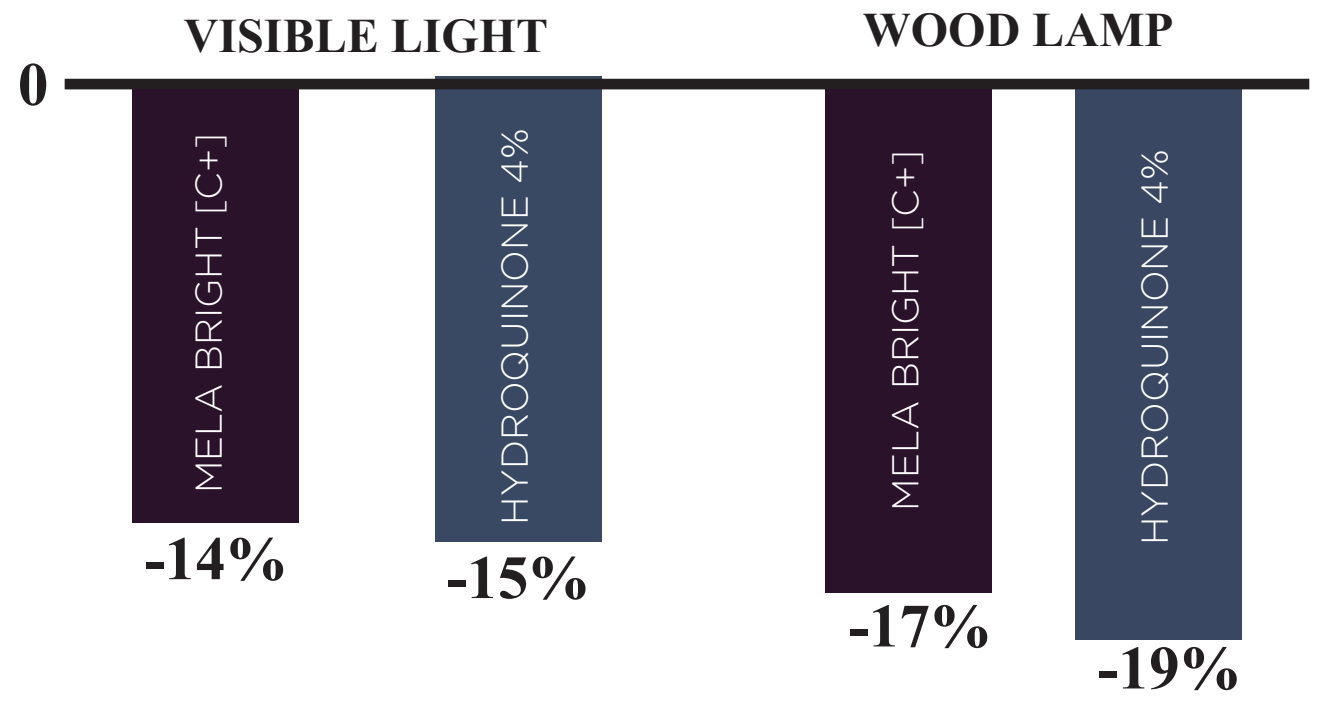
VISIBLE LIGHT
WOOD LAMP
0
MELA BRIGHT [C+]
HYDROQUINONE 4%
MELA BRIGHT [C+]
HYDROQUINONE 4%
-14%
-15%
-17%
-19%

Pigmentation intensity evolution after 2 months, measured by VISIA®

MASI score

The evaluation is on-going for 60 additional days, the MASI scores will be calculated at the end of the study at 120 days.

SKIN TOLERANCE AND SIDE EFFECTS

MELA BRIGHT® [C+] showed an excellent skin tolerance profile, with no drop-out and no skin irritation reported during the study.

CASES

In the best cases, a reduction of pigmentation intensity up to 84% (Wood lamp) and 52% (visible light) on the MELA BRIGHT® [C+] side was measured.

T0 **T2 months**

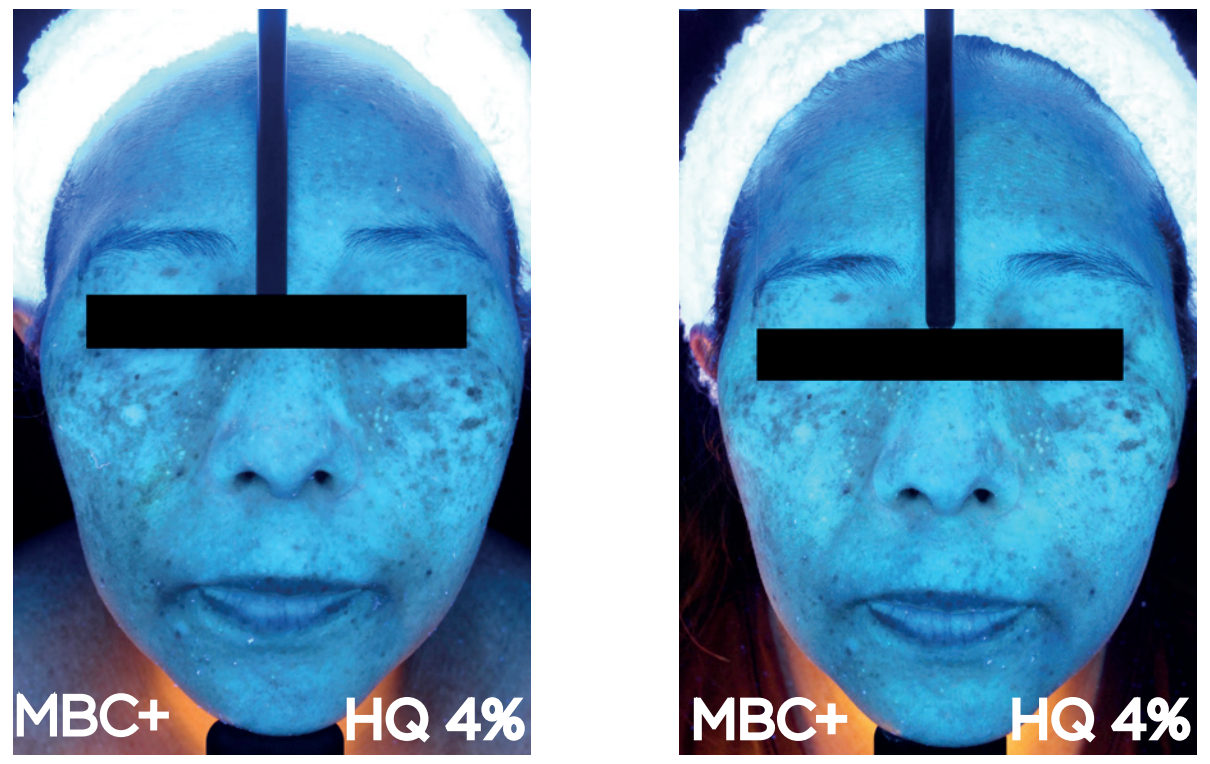
MBC+
HQ 4%
MBC+
HQ 4%

Case n°5 – Melasma - Wood lamp

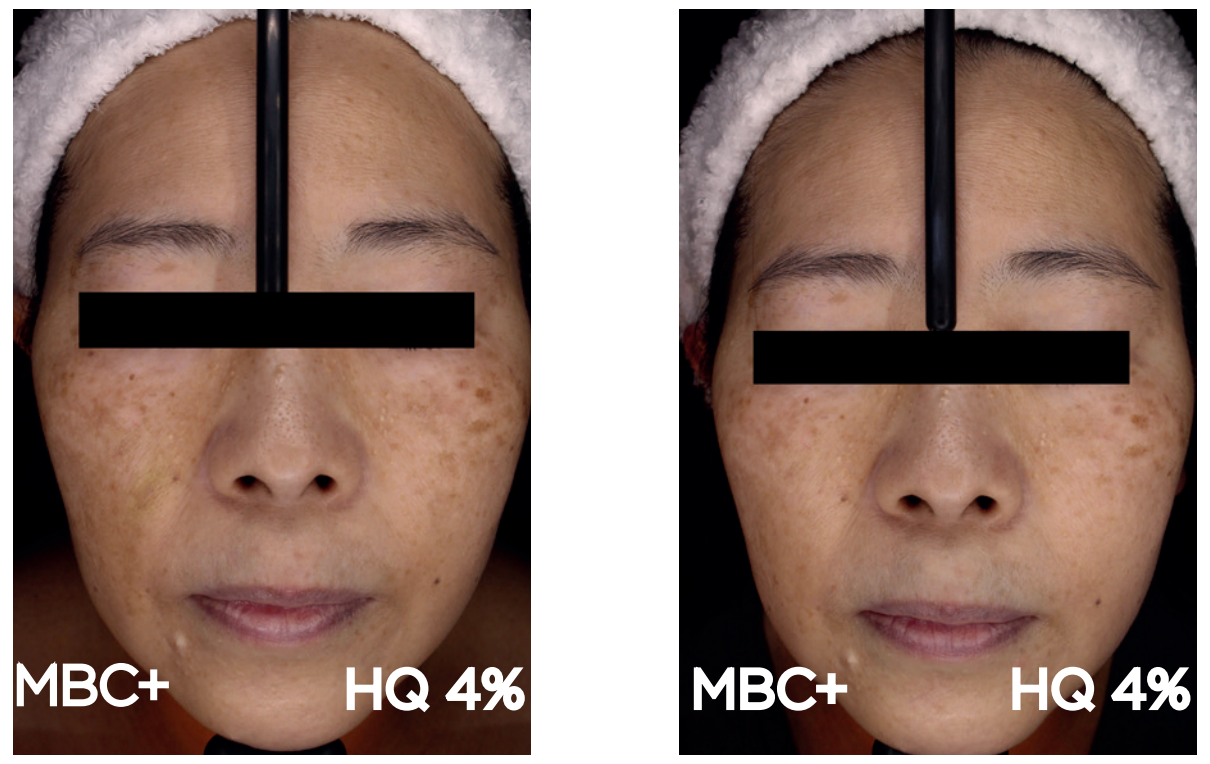
MBC+
HQ 4%
MBC+
HQ 4%

Case n°5 – Melasma - visible light

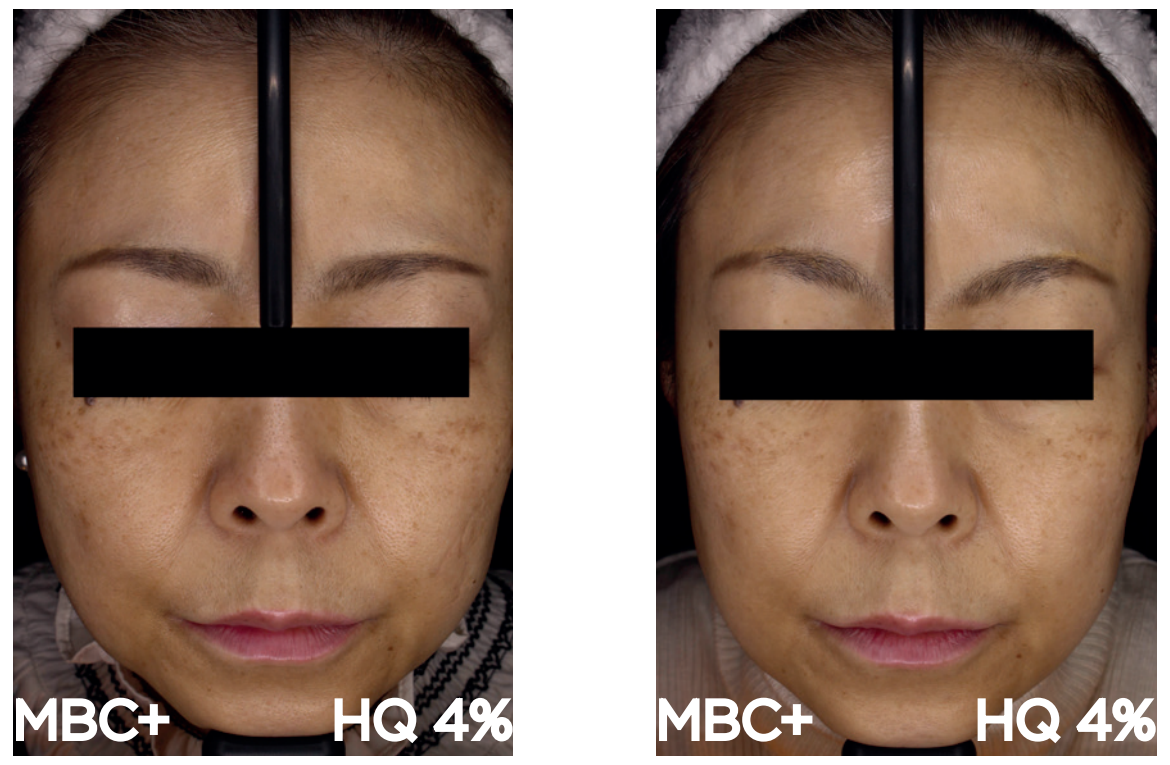
MBC+
HQ 4%
MBC+
HQ 4%

Case n°21 – Melasma - visible light

REDNESS

A decrease of 2% in redness was measured in the the MELA BRIGHT® [C+] side, compared to an increase of 1% in the HYDROQUINONE 4% side. These results are not statistically significant.